476LB

Zelecimient Rostudirsen Led to Trends in Long-term Improvement in Clinical Outcomes Including Cardiopulmonary Function: Additional Data from DELIVER

Chamindra G. Laverty[1*], Susan Apkon[2], Liesbeth De Waele[3], Kevin M. Flanigan[4], Hugh McMillan[5], Marika Pane[6], Perry Shieh[7], Ian R. Woodcock[8], Prashant Bansal[9], Soma Ray[9], Dazhe Wang[9], Douglas Kerr[9], Maria L. Naylor[9], Michela Guglieri[10] on behalf of the DELIVER study investigators.

*Presenting author.
[1]UC San Diego Health San Diego, CA, USA; [2]Children's Hospital Colorado, Aurora, CO, USA; [3]University Hospitals Leuven, Leuven, Belgium; [4]Nationwide Children's Hospital, Columbus, OH, USA;
[5]Children's Hospital of Eastern Ontario, Ottawa, Canada; [6]Fondazione Policlinico Universitario A. Gemelli, Rome, Italy; [7]University of California, Los Angeles, Los Angeles, CA, USA;
[8]Murdoch Children's Research Institute, Parkville, Victoria and The Royal Children's Hospital, Melbourne, Australia; [9]Dyne Therapeutics, Inc., Waltham, MA, USA; [10]Royal Victoria Infirmary, Newcastle University, Newcastle-Upon-Tyne, UK.

View on the Dyne Therapeutics publications website:

DELIVER

BACKGROUND

- Duchenne muscular dystrophy (DMD) is a rare, X-linked, recessive, progressive neuromuscular disorder in which mutations in the *DMD* gene lead to a greatly reduced or absent dystrophin protein which is essential for muscle structure, function, and preservation[1–4]
- DMD is a multisystem disease that affects skeletal, including pulmonary, smooth, and cardiac muscles as well as the central nervous system (CNS), and individuals with DMD typically exhibit progressive muscle weakness followed by loss of ambulation and cardiopulmonary failure, the leading cause of mortality[2,5,6]
- Zeleciment rostudirsen (z-rostudirsen, also known as DYNE-251) leverages transferrin receptor 1 (TfR1) to deliver an exon 51 skipping phosphorodiamidate morpholino oligomer (PMO) to key tissues relevant to DMD, with the goal of producing near-full-length, functional dystrophin[7,8]
- The safety and efficacy of z-rostudirsen are being investigated in the Phase 1/2 DELIVER trial (NCT05524883; EU Trial [CTIS] number 2023-510351-31-00)[9]
- DELIVER is a global, double-blind, randomized, placebo-controlled study evaluating intravenous male participants with DMD (4–16 years old) with mutations amenable to exon 51-skipping therapy administrations of z-rostudirsen in ambulant and non-ambulant (Figure 1)[10]
 - At 6 months, functional improvement was observed across multiple measures of muscle function relative to baseline and placebo, including time-to-rise (TTR) velocity, 10-meter walk/run (10MWR) velocity, North Star Ambulatory Assessment (NSAA), stride velocity 95th centile (SV95C), performance upper limb v2.0 (PUL2.0), and forced vital capacity percent predicted (FVC%p)[10]

METHODS

- DELIVER consists of a multiple ascending dose (MAD) portion to identify the optimal dose regimen of z-rostudirsen, and a registrational expansion cohort (REC) to evaluate the safety and efficacy of the selected dose (20 mg/kg Q4W) in participants with DMD mutations amenable to exon 51 skipping. After the 24-week placebo-controlled period in the MAD and REC, participants entered an open-label extension/long-term extension (OLE/LTE) and transitioned to 20 mg/kg Q4W, as applicable (Figure 1)
- A total of 86 participants have been enrolled in DELIVER
- Here, we present long-term efficacy data for a subset of participants from the DELIVER study up to 24 months, as well as safety data for all 86 participants up to 36 months of follow-up
- To characterize the natural history of pulmonary and cardiac outcomes in individuals with DMD, 2-year trendlines were derived from published literature
 - Estimated trendlines for pulmonary function assume an average annual decline of ~5%, based on Meier et al (2017),[11] Mayer et al (2015),[12] and McDonald et al (2018),[13] with a mean baseline age >10 years
 - Estimated trendlines for left ventricular ejection fraction (LVEF) and circumferential strain assume an average annual decline of ~1%, based on Batra et al (2022)[14] and Hagenbuch et al (2010),[15] with a mean baseline age >11 years

Figure 1. DELIVER study design

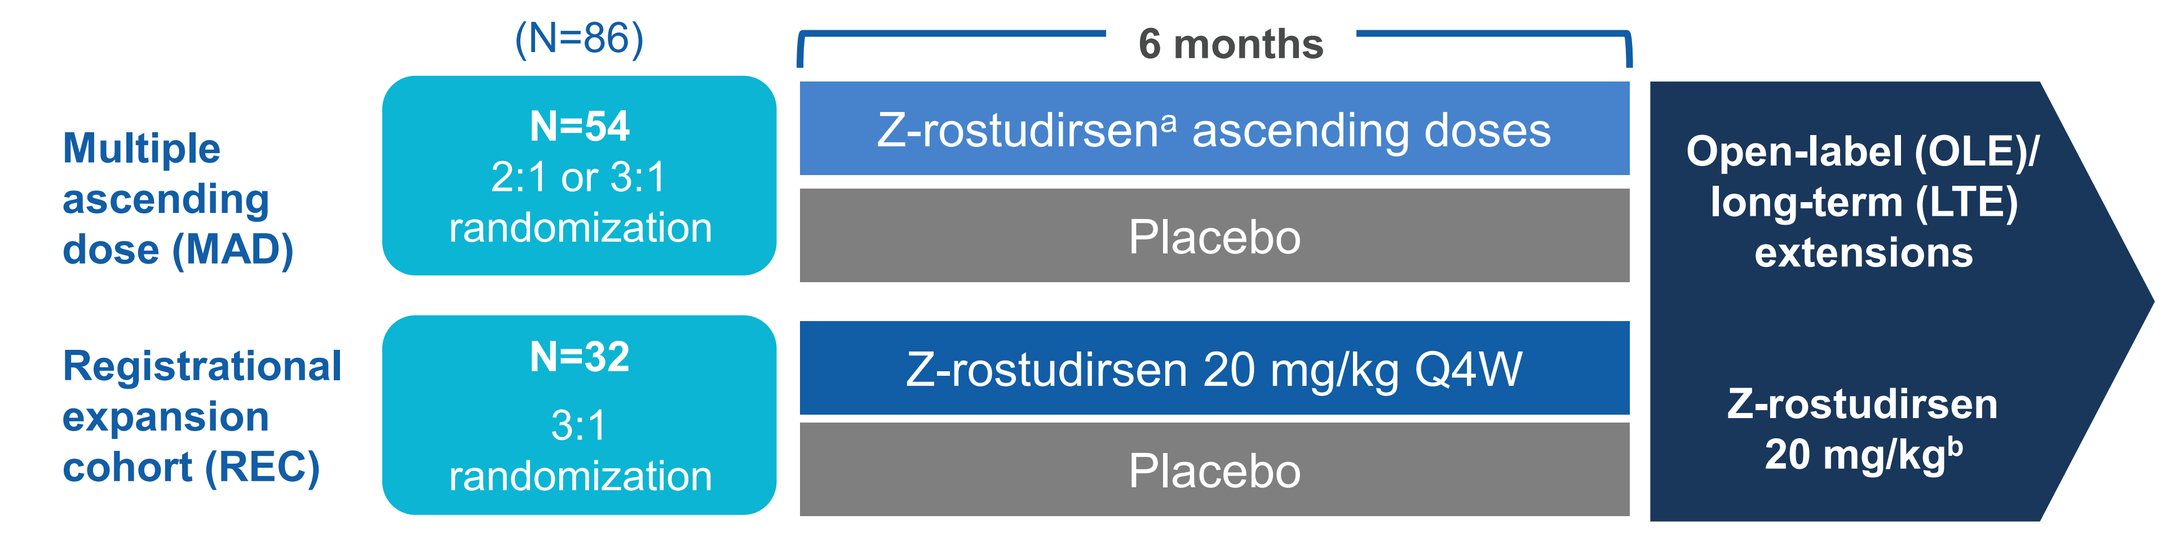

(N=86)
6 months
Multiple ascending dose (MAD)
N=54
2:1 or 3:1 randomization
Z-rostudirsen[a] ascending doses
Placebo
Registrational expansion cohort (REC)
N=32
3:1 randomization
Z-rostudirsen 20 mg/kg Q4W
Placebo
Open-label (OLE)/ long-term (LTE) extensions
Z-rostudirsen 20 mg/kg[b]

Select inclusion/exclusion criteria
- Inclusion: Ambulatory or non-ambulatory; age 4 to 16 years inclusive; stable dosage of glucocorticoids for at least 12 weeks
- Exclusion: Exon-skipping/dystrophin-modifying therapy or givinostat within 12 weeks of randomization; gene therapy at any time

Primary endpoints: Change from baseline in dystrophin protein levels by Western blot; safety and tolerability

Key functional endpoints: TTR velocity, 10MWR velocity, NSAA, SV95C, PUL2.0, FVC%p

a. Z-rostudirsen doses in the MAD cohorts ranged from 0.7 mg/kg to 40 mg/kg every 4 or 8 weeks; b. Transition to 20 mg/kg dose occurred at non-uniform times during OLE or LTE; for participants initiated at 40 mg/kg, transition started either in the placebo-controlled period or OLE.
10MWR, 10-meter walk/run; FVC%p, forced vital capacity percent predicted; NSAA, North Star Ambulatory Assessment; PUL2.0, performance upper limb v2.0; Q4W, every 4 weeks; SV95C, stride velocity 95th centile; TTR, time to rise.

RESULTS

Table 1. Baseline characteristics

Mean (SD) or n (%)	Placebo (MAD+REC) N=24[e]	Z-rostudirsen 10 → 20 mg/kg[f] Q4W (MAD) N=6	Z-rostudirsen 20 mg/kg Q4W (MAD) N=6	Overall (MAD+REC) N=86
Age (years)	8.2 (2.5)	6.8 (2.5)	7.7 (2.5)	8.3 (3.3)
BMI (kg/m^2)	19.8 (4.7)	17.9 (3.7)	17.5 (2.9)	18.7 (4.3)
Age of symptom onset (years)	3.4 (1.8)	3.0 (1.8)	2.0 (0.9)	2.8 (1.7)
Most recent corticosteroid dosing regimen, n (%)[a]				
Daily	20 (83.3)	6 (100)	6 (100)	73 (84.9)
Other	4 (16.7)	0	0	13 (15.1)
Duration of corticosteroid treatment (years)[b]	2.1 (2.4)	1.5 (2.0)	2.4 (2.2)	2.6 (2.6)
Prior DMD therapy, n (%)				
Eteplirsen	4 (16.7)	0	0	15 (17.4)
Other	2 (8.3)	0	2 (33.3)	15 (17.4)
PUL2.0 total score[c]	36.3 (4.0)	37.2 (5.9)	33.8 (3.5)	36.4 (4.2)
FVC%p	92.7 (17.6)	89.8 (22.7)	90.7 (11.2)	87.5 (19.2)
Ambulant (%)	19 (79.2)	5 (83.3)	6 (100)	75 (87.2)
TTR velocity (rise/sec)[d]	0.20 (0.10)	0.23 (0.06)	0.17 (0.14)	0.20 (0.1)
10MWR velocity (m/sec)[d]	2.0 (0.5)	2.5 (0.7)	1.5 (0.6)	1.9 (0.6)
NSAA total score[d]	21.6 (6.3)	26.6 (5.4)	15.0 (5.3)	20.6 (7.0)
SV95C (m/sec)[d]	1.7 (0.5)	2.0 (0.3)	1.4 (0.5)	1.6 (0.5)

a. Most recent corticosteroid regimen refers to corticosteroid at time of randomization. b. Cumulative duration of previous and most recent corticosteroid treatment at the time of randomization. c. Missing values imputed. d. Ambulant participants; out-of-threshold and/or missing values imputed. e. All placebo participants pooled from MAD and REC. f. Participants transitioned from 10 mg/kg Q4W to 20 mg/kg Q4W after 6 months. 10MWR, 10-meter walk/run; BMI, body mass index; DMD, Duchenne muscular dystrophy; FVC%p, forced vital capacity percent predicted; m, meter; MAD, multiple ascending dose; NSAA, North Star Ambulatory Assessment; PUL2.0, performance upper limb v2.0; SD, standard deviation; Q4W, every 4 weeks; REC, registrational expansion cohort; SD, standard deviation; sec, second; SV95C, stride velocity 95th centile; TTR, time to rise.

Figure 2. Z-rostudirsen led to sustained improvement in multiple measures of muscle function through 24 months

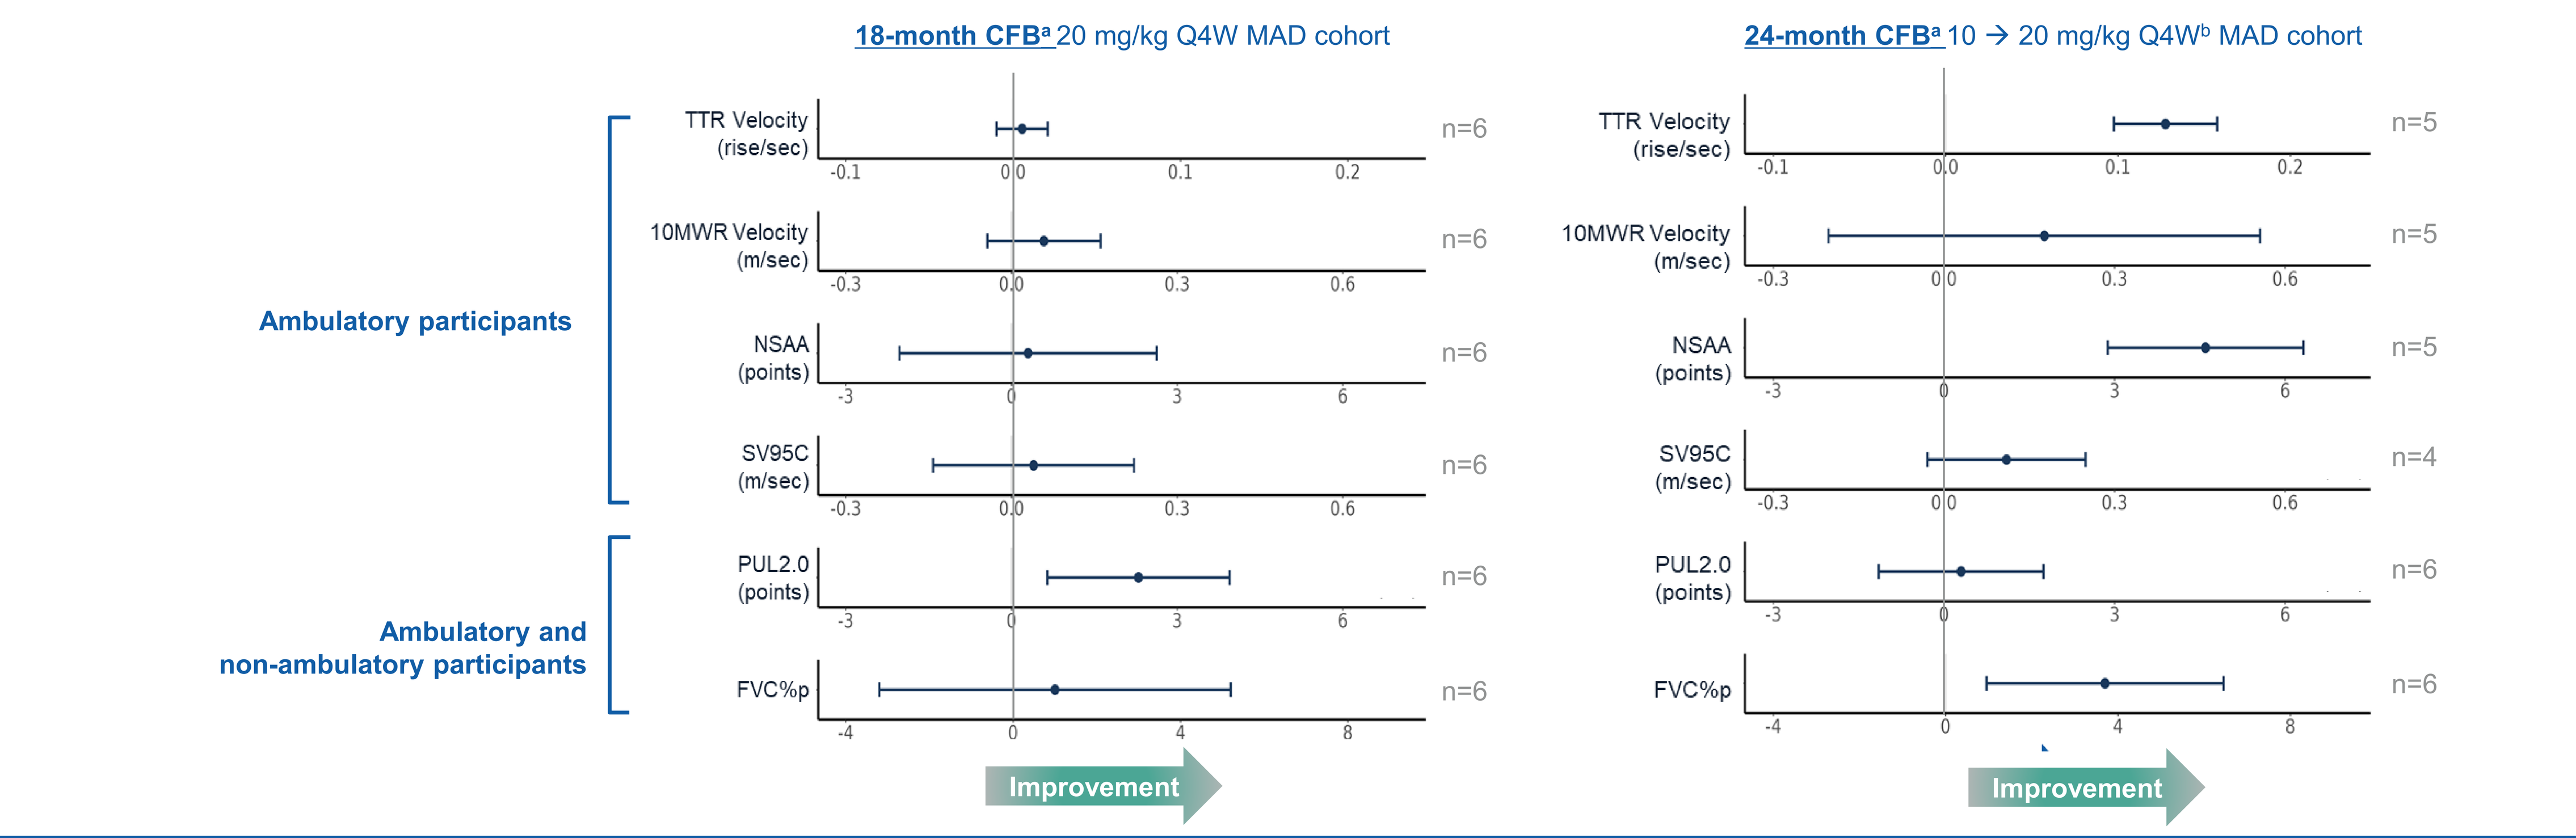

18-month CFB[a] 20 mg/kg Q4W MAD cohort
24-month CFB[a] 10 → 20 mg/kg Q4W[b] MAD cohort
Ambulatory participants
Ambulatory and non-ambulatory participants
TTR Velocity (rise/sec)
10MWR Velocity (m/sec)
NSAA (points)
SV95C (m/sec)
PUL2.0 (points)
FVC%p
n=6
n=5
n=4
Improvement

a. Mean CFB +/- SEM; TTR velocity, 10MWR velocity, NSAA, and SV95C analyzed from ambulant participants; PUL2.0 and FVC%p analyzed from ambulant and non-ambulant participants; Out-of-threshold and/or missing values imputed except for FVC%p. b. Participants transitioned from 10 mg/kg Q4W to 20 mg/kg Q4W after 6M; all participants treated with 20 mg/kg Q4W for at least 12M in the 24M assessment.
10MWR, 10-meter walk/run; CFB, change from baseline; FVC%p, forced vital capacity percent predicted; m, meters; M, months; MAD, multiple ascending dose; NSAA, North Star Ambulatory Assessment; PUL2.0, performance upper limb v2.0; Q4W, every 4 weeks; s, second; SV95C, stride velocity 95th centile; SEM, standard error of mean; TTR, time to rise.

Figure 3. Improvement from baseline in lung function contrasts with an expected decline based on published natural history data

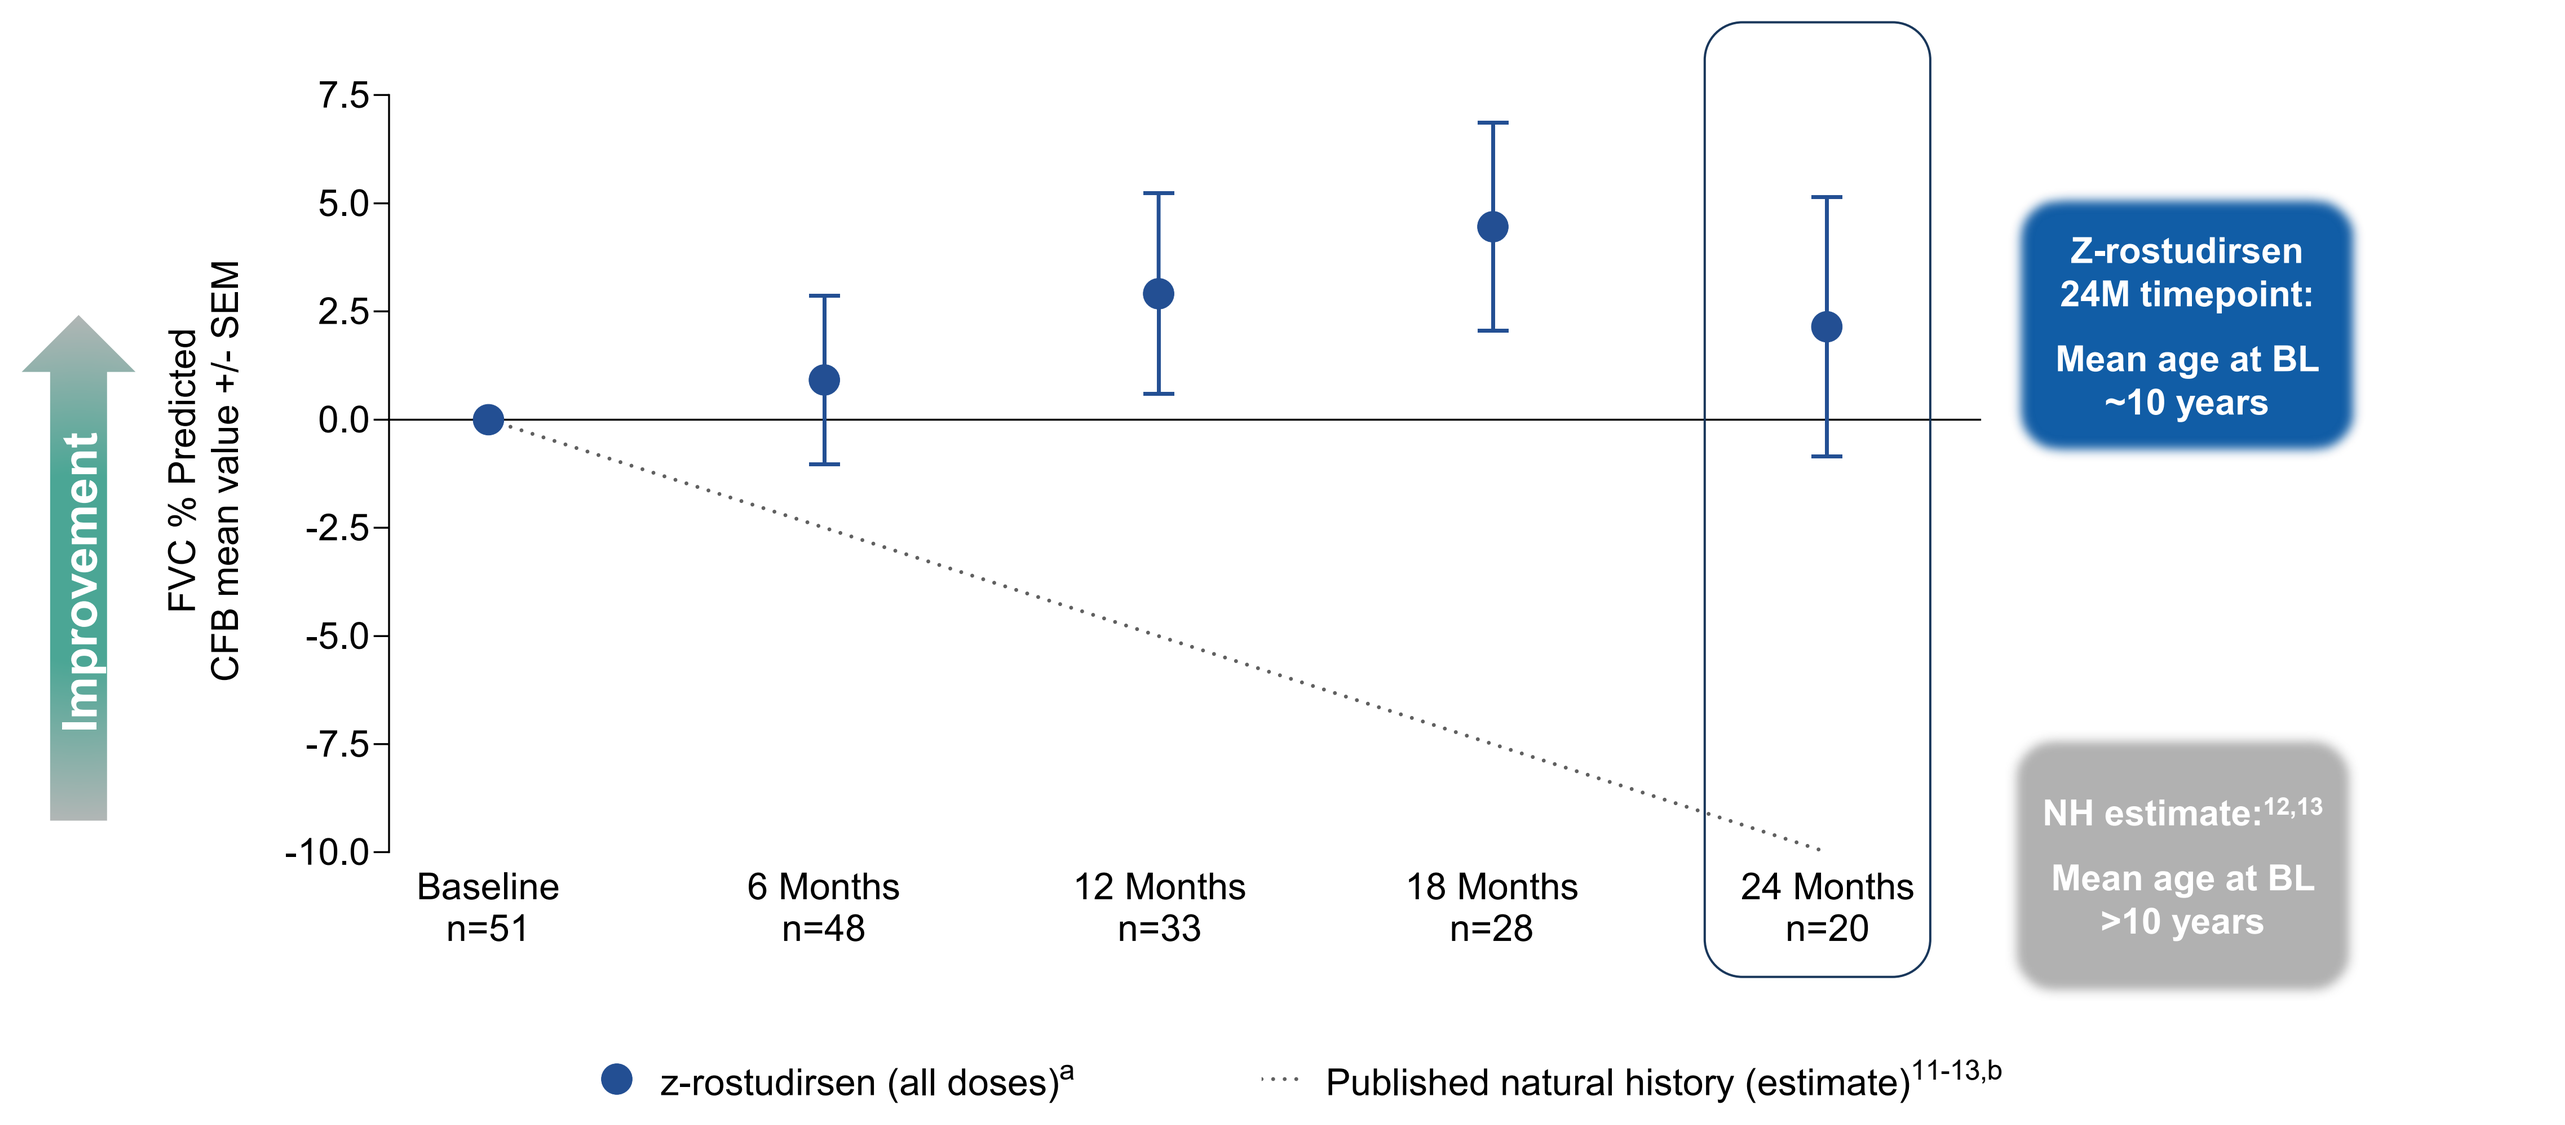

Improvement
FVC % Predicted CFB mean value +/- SEM
Baseline n=51
6 Months n=48
12 Months n=33
18 Months n=28
24 Months n=20
Z-rostudirsen 24M timepoint:
Mean age at BL ~10 years
NH estimate:[12,13]
Mean age at BL >10 years
z-rostudirsen (all doses)[a]
Published natural history (estimate)[11-13,b]

- The outcomes represent a mixture of exposures and do not reflect the effect of maintaining 20 mg/kg for the entire treatment duration
- The majority of participants at the 24M timepoint initiated treatment at the 0.7–2.8 mg/kg Q4W dose levels. One participant initiated treatment at 20 mg/kg Q4W. All participants at the 24M timepoint received 20 mg/kg Q4W z-rostudirsen for ≥9 months

Results are reported as mean CFB through 24 months and presented with SEM.
a. Shown are all DELIVER participants (REC + MAD) randomized to z-rostudirsen treatment at baseline (any dose, including dose escalation[s] or de-escalation) and for whom FVC%p data were available. b. Based on 2-year published natural history data; assumes ~5%/year decline in FVC%p. (Meier 2017; Mayer 2015; Macdonald 2018).
BL, baseline; CFB, change from baseline; FVC%p, forced vital capacity percent predicted; M, month; MAD, multiple ascending dose; NH, natural history; REC, registrational expansion cohort; SEM, standard error of the mean.

Figure 4: Trend of sustained improvement in CMR-based circumferential strain, an early signal of preserved myocardial contractile quality

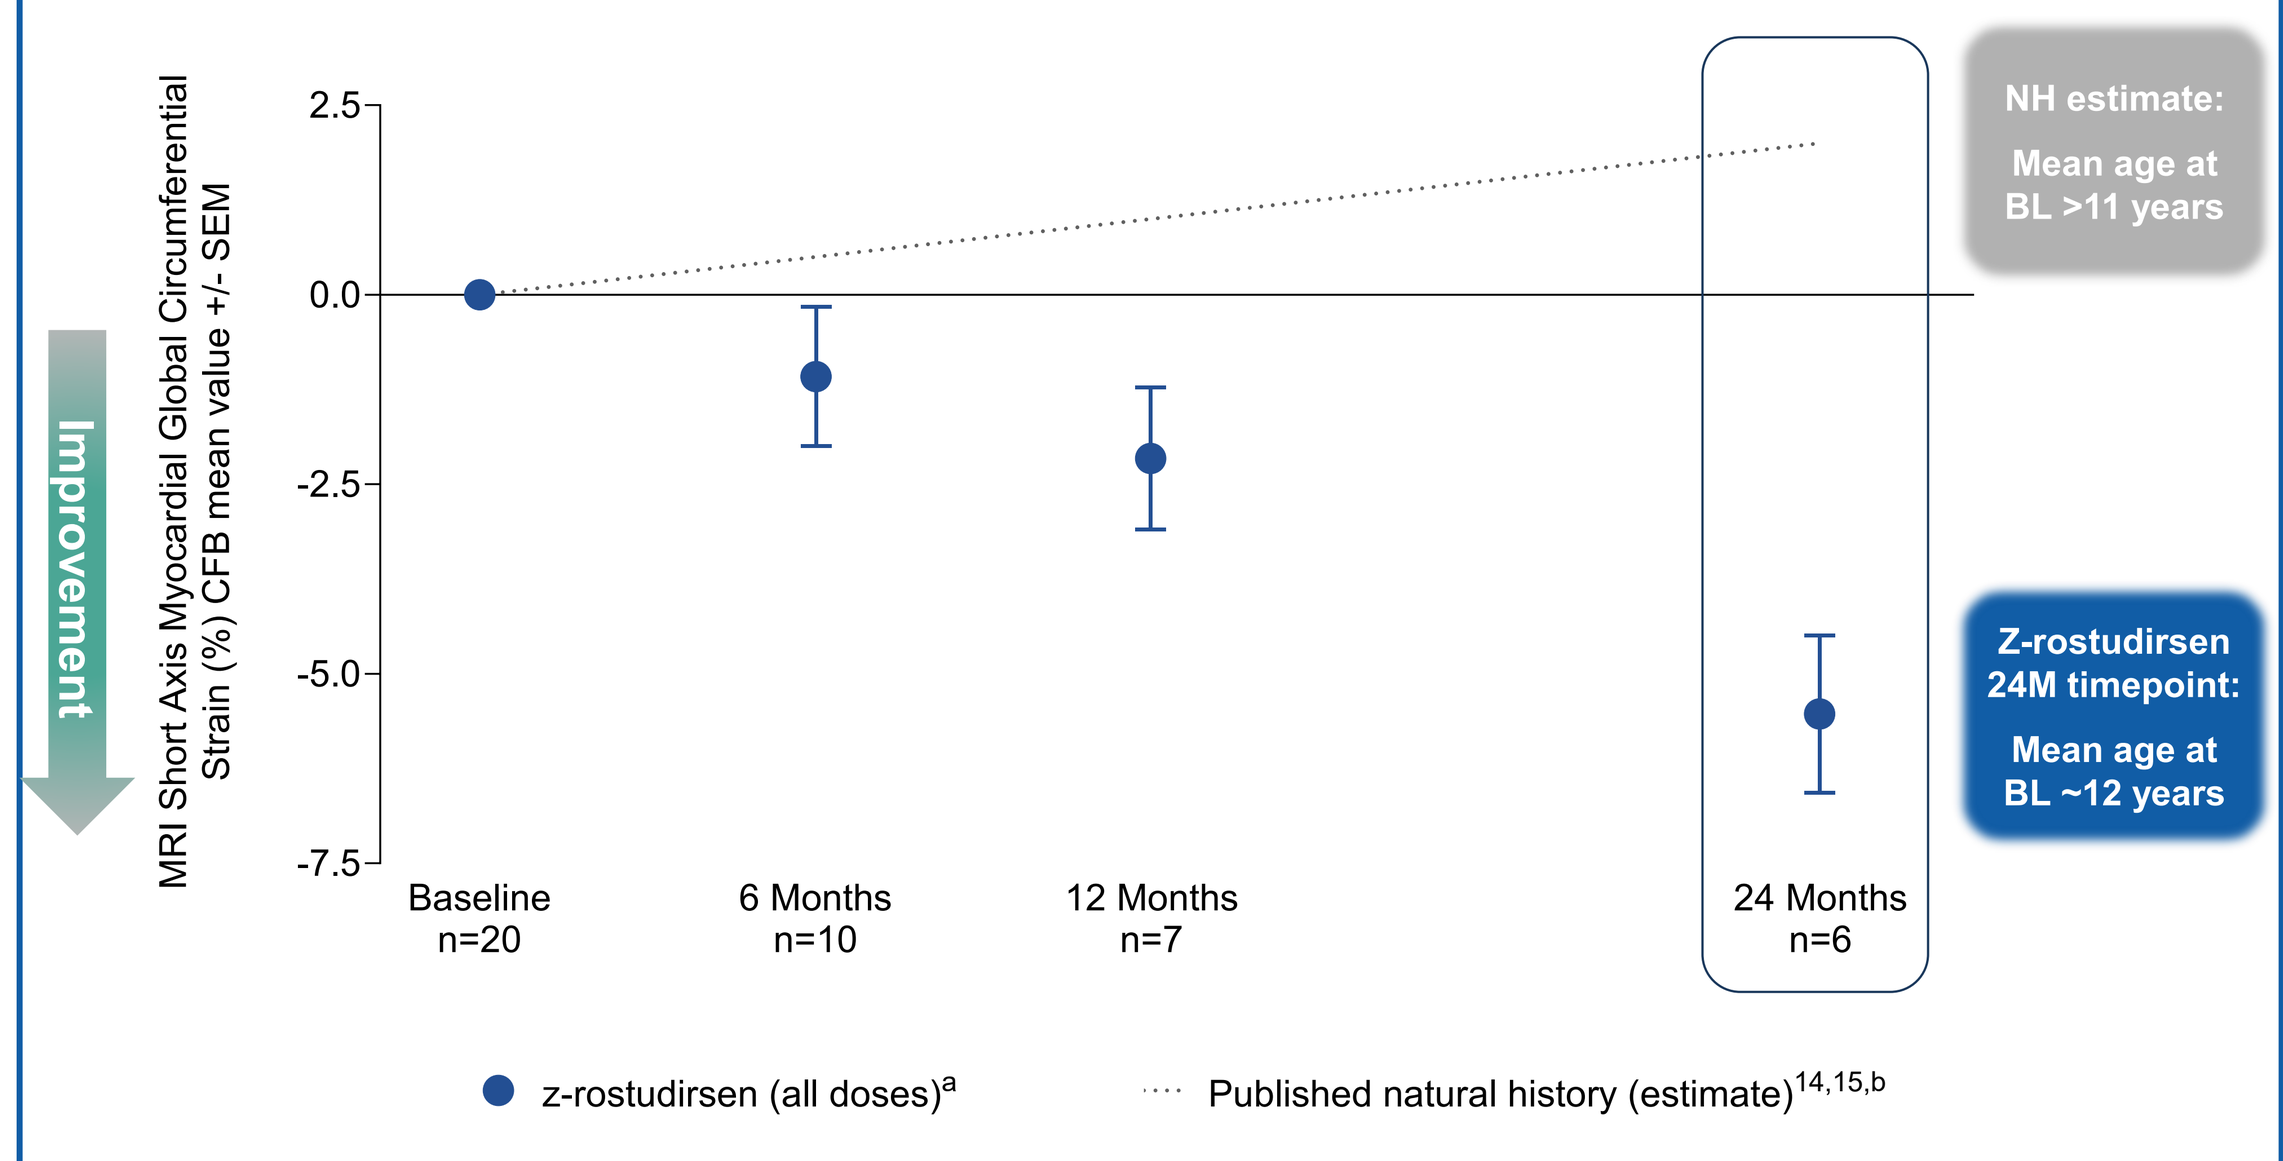

Improvement
MRI Short Axis Myocardial Global Circumferential Strain (%) CFB mean value +/- SEM
Baseline n=20
6 Months n=10
12 Months n=7
24 Months n=6
NH estimate:
Mean age at BL >11 years
Z-rostudirsen 24M timepoint:
Mean age at BL ~12 years
z-rostudirsen (all doses)[a]
Published natural history (estimate)[14,15,b]

- The outcomes represent a mixture of exposures and do not reflect the effect of maintaining 20 mg/kg for the entire treatment duration
- The majority of participants at the 24M timepoint initiated treatment at the 0.7–2.8 mg/kg Q4W dose levels. One participant initiated treatment at 20 mg/kg Q4W. All participants at the 24M timepoint received 20 mg/kg Q4W z-rostudirsen for ≥6 months

Results are reported as mean CFB through 24 months and presented with SEM.
a. Shown are all DELIVER participants (REC + MAD) randomized to z-rostudirsen treatment at baseline (any dose, including dose escalation[s] or de-escalation) and for whom circumferential strain data were available. b. Based on 2-year published natural history data; assumes ~1%/year decline in strain (Batra 2022; Hagenbuch 2010). BL, baseline; CFB, change from baseline; CMR, cardiac magnetic resonance; M, month; MAD, multiple ascending dose; MRI, magnetic resonance imaging; NH, natural history; REC, registrational expansion cohort; SEM, standard error of the mean.

Figure 5. Positive trend in CMR-based left ventricular ejection fraction contrasts an expected decline based on published natural history data

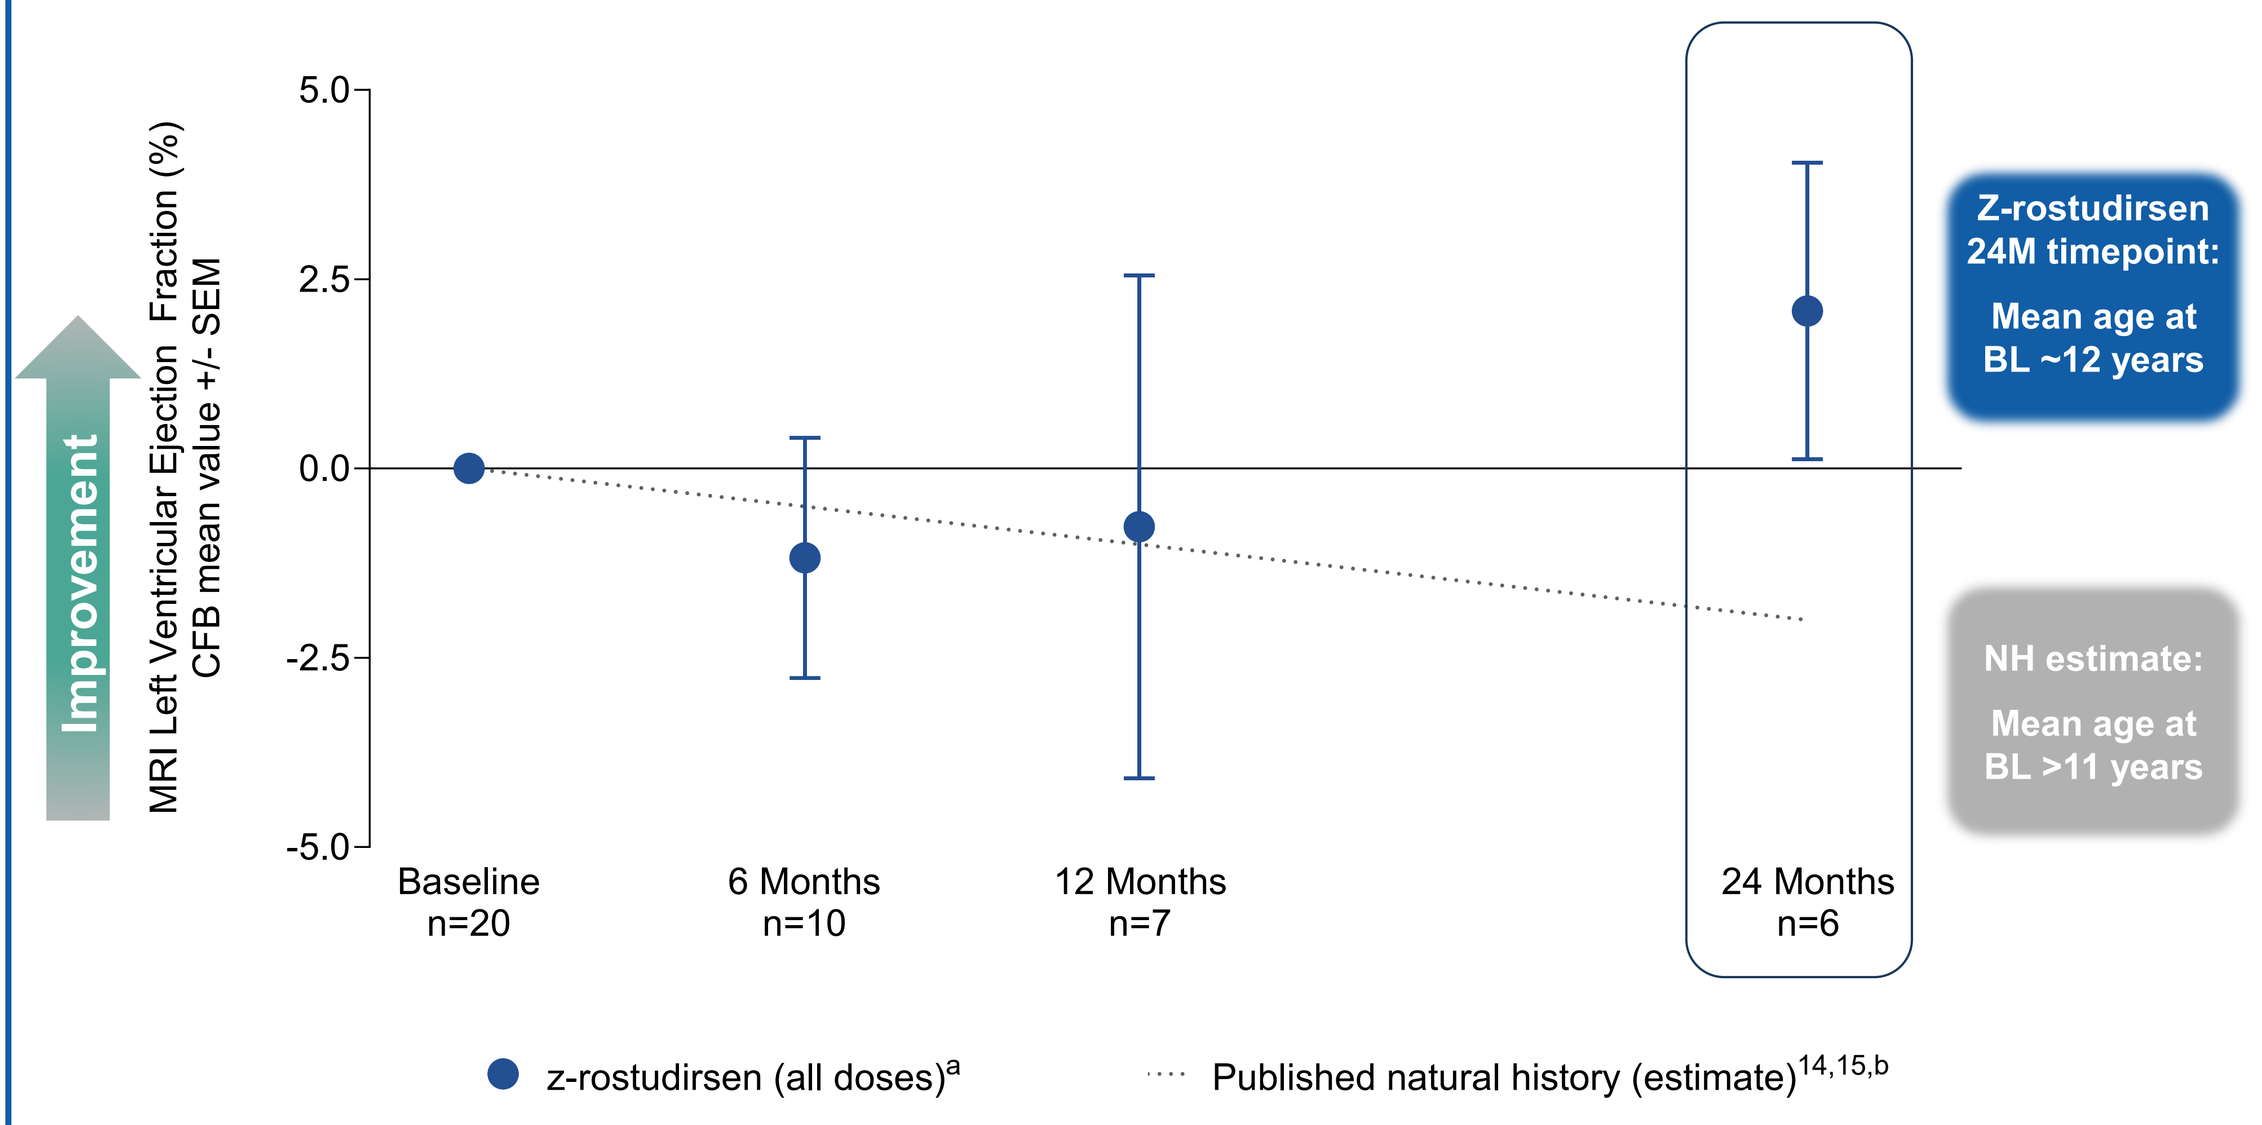

Improvement
MRI Left Ventricular Ejection Fraction (%) CFB mean value +/- SEM
Baseline n=20
6 Months n=10
12 Months n=7
24 Months n=6
Z-rostudirsen 24M timepoint:
Mean age at BL ~12 years
NH estimate:
Mean age at BL >11 years
z-rostudirsen (all doses)[a]
Published natural history (estimate)[14,15,b]

- The outcomes represent a mixture of exposures and do not reflect the effect of maintaining 20 mg/kg for the entire treatment duration
- The majority of participants at the 24M timepoint initiated treatment at the 0.7–2.8 mg/kg Q4W dose levels. One participant initiated treatment at 20 mg/kg Q4W. All participants at the 24M timepoint received 20 mg/kg Q4W z-rostudirsen for ≥6 months

Results are reported as mean CFB through 24 months and presented with SEM.
a. Shown are all DELIVER participants (REC + MAD) randomized to z-rostudirsen treatment at baseline (any dose, including dose escalation[s] or de-escalation) and for whom LVEF data were available.
b. Based on 2-year published natural history data; assumes ~1%/year decline in LVEF (Batra 2022; Hagenbuch 2010).
BL, baseline; CFB, change from baseline; CMR, cardiac magnetic resonance; LVEF, left ventricular ejection fraction; M, month; MAD, multiple ascending dose; MRI, magnetic resonance imaging; NH, natural history; REC, registrational expansion cohort; SEM, standard error of the mean.

Table 2. Safety profile of z-rostudirsen 20 mg/kg Q4W remains favorable: Summary of TEAEs[a]

Study period	Placebo-controlled period (0 to 6M)		All study periods (0 to ≤36M)
Participants with ≥1 TEAE – n (%)	Placebo (MAD+REC) N=24[b]	Z-rostudirsen 20 mg/kg Q4W (MAD+REC) N=30[c]	Z-rostudirsen pooled doses[d] (MAD+REC) N=85[e]
Any TEAE	22 (91.7)	29 (96.7)	80 (94.1)
Any related TEAE	3 (12.5)	10 (33.3)	41 (48.2)
Any serious TEAE	1 (4.2)	2 (6.7)	10 (11.8)
Any serious related TEAE	0	0	4 (4.7)
Any TEAE leading to withdrawal from study	0	0	0
Any TEAE leading to death	0	0	0

- Most related TEAEs were mild or moderate
- Potentially related serious TEAEs
 - 2 participants at 20 mg/kg Q4W (registrational dose)
 - Pyrexia (fever) and malaise[f]
 - 2 participants at 40 mg/kg Q4W
 - Acute kidney injury; thrombocytopenia[g]
 - Pancytopenia[h]
- Most frequent related TEAEs ≥10%[i]
 - Pyrexia (fever) (18%)
 - Headache (13%)
- Additional safety data at 20 mg/kg Q4W
 - No participants have persistent related anemia[j] or thrombocytopenia

1441 doses of z-rostudirsen administered to date representing 113 patient-years of follow-up (up to 36 months).[a] 1062 doses of z-rostudirsen at 20 mg/kg dose level administered to date.[a]

a. Data as of August 19, 2025; all participants, placebo-controlled period, OLE, and LTE. b. All placebo participants pooled from MAD and REC. c. All participants randomized to z-rostudirsen 20 mg/kg Q4W in MAD and REC cohorts. d. All doses of z-rostudirsen from MAD and REC at doses ranging from 0.7 mg/kg to 40 mg/kg every 4 or 8 weeks. e. One participant randomized to placebo in REC not yet dosed with z-rostudirsen as of August 19, 2025. f. One participant with same-day onset of pyrexia and malaise in OLE and separate single event of pyrexia in LTE; one participant with single event of pyrexia in LTE; both participants fully recovered and have continued to receive z-rostudirsen without interruption. g. Events had same day of onset in a single participant with a non-serious related TEAE of anemia in the context of fever, hemolysis, diarrhea, and positive blood in stool; together these events were consistent with hemolytic uremic syndrome with a possible infectious etiology. h. Participant has a history of hemolytic anemia of unidentified etiology; presented with fever and tonsilitis; symptoms resolved without therapeutic intervention. i. All cohorts combined; preferred terms reported. j. No participants have persistent related anemia with Hgb levels <11.2 g/dL (threshold for anemia in children[16]).
Hgb, hemoglobin; LTE, long-term extension; M, months; MAD, multiple ascending dose; OLE, open-label extension; Q4W, every 4 weeks; REC, registrational expansion cohort; TEAE, treatment-emergent adverse event.

CONCLUSIONS

- Long-term data from the DELIVER trial demonstrated multisystem functional improvement across upper and lower limb, trunk, cardiac and pulmonary muscles
- In participants who were randomized to receive z-rostudirsen at baseline (across all dose cohorts), there were trends in improvement from baseline in both lung and cardiac function through 24 months, in contrast to the expected declines based on published natural history in DMD
- Z-rostudirsen demonstrated a favorable safety and tolerability profile based on data from 86 participants enrolled in DELIVER and followed for up to 36 months[a]
- These long-term data from the DELIVER trial support the potential of z-rostudirsen to address the unmet needs of individuals with DMD amenable to exon 51 skipping

a. As of August 19, 2025.

DISCLOSURE INFORMATION

Chamindra G. Laverty has received consulting fees from Sarepta, Genentech, and Italfarmaco; Susan Apkon is a site PI for Dyne Therapeutics, Capricor, Sarepta, Satellos, Biohaven, ScholarRock, and Biogen; Liesbeth De Waele is PI on studies sponsored by Sarepta Therapeutics, Italfarmaco, Pfizer and Dyne Therapeutics and has participated in ad hoc advisory board activities for Santhera, Pfizer and Italfarmaco; Kevin Flanigan has received clinical trial support from Sarepta, Dyne Therapeutics, Avidity, Ultragenyx, and Solid. He has served on advisory boards for Armatus, Encoded, Insmed, Dyne Therapeutics, Solid, Precision Biosciences, and received consultation fees from Satellos; Hugh McMillan has received consulting fees from Roche, Novartis, Key Pharma, Regenxbio, Solid Biosciences, and Merck; Marika Pane has nothing to declare; Perry Shieh is a consultant for Sarepta Therapeutics, Dyne Therapeutics, Biogen, Genentech, Novartis, Astellas, Solid, Sanofi, Alexion, Argenx, CSL Behring, Grifols and UCB and has received research grants from Sarepta Therapeutics, Solid Biosciences, PTC, Dyne Therapeutics, Biogen, Genentech, Novartis, Astellas, Avidity Biosciences, AMO Pharma, Abcuro and Sanofi; Ian R Woodcock has received honoraria for work performed including educational activities and attendance at advisory board meetings from pharmaceutical companies Avidity, Biogen, Juvena Therapeutics, Keros Therapeutics, Novartis, Pfizer, Roche, Solid Biosciences and Soufflé Therapeutics. He has received grants for research work from Fulcrum Therapeutics, FSHD Global Research Foundation, FSHD Society, NIH and has been PI on a number of industry-sponsored clinical trials, including those sponsored by Avidity, Biogen, Catabasis, Dyne Therapeutics, EryDel, Keros Therapeutics, Novartis, Pfizer, Percheron Therapeutics, PTC, Roche, Sarepta, Satellos, and Solid Biosciences. None of these disclosures affected the work he performed on this work; Michela Guglieri chaired a study sponsored by ReveraGen (no financial benefits) and had research collaborations with ReveraGen and Sarepta Therapeutics. She acted as CI/PI for clinical trials sponsored by Dyne Therapeutics, Pfizer, Italfarmaco, Edgewise, Roche, Santhera, ReveraGen and Dynacure, and participated in advisory boards for Pfizer, NS Pharma, Dyne Therapeutics (consultancies through Newcastle University). She has received speaker honoraria from Sarepta Therapeutics, Italfarmaco and Novartis; Prashant Bansal, Soma Ray, Dazhe Wang, Douglas Kerr and Maria L. Naylor are current or former employees of Dyne Therapeutics and may hold stock in the company.

ACKNOWLEDGMENTS

This study was funded by Dyne Therapeutics. Medical writing and editorial support was provided by Obsidian Healthcare Group Ltd, UK. This support was funded by Dyne Therapeutics. The authors wish to thank the clinical trial investigators and site coordinators and, most importantly, all the participants, families, and caregivers for their willingness to participate in this clinical trial, which was sponsored by Dyne Therapeutics.

REFERENCES

1. Crisafulli S, et al. *Orphanet J Rare Dis.* 2020;15(1):141;
2. Birnkrant DJ, et al. *Lancet Neurol.* 2018;17(3):251–267;
3. de Feraudy Y, et al. *Ann Neurol.* 2021;89(2):280–292;
4. Claflin DR, Brooks SV. *Am J Physiol Cell Physiol.* 2008;294(2):C651–C658;
5. Ohlendieck K, Swandulla D. *Pflügers Arch.* 2022;473(12):1813–1839;
6. Birnkrant DJ, et al. Lancet Neurol. 2018;17(4):347–361;
7. Desjardins CA, et al. *Nucleic Acids Res.* 2022;50(20):11401–11414;
8. Zanotti, S. Oral presentation at the 26th American Society of Gene and Cell Therapy Annual Meeting. May 17, 2023. Los Angeles, USA. Abstract 82.
9. ClinicalTrials.gov. NCT05524883. Available from: https://clinicaltrials.gov/study/NCT05524883. Accessed February 23, 2026;
10. Flanigan K, et al. Annual MDA Clinical & Scientific Conference; Orlando, FL; Mar 8–11, 2026. Poster 442O;
11. Meier T, et al. *Neuromuscul Disord.* 2017;27(4):307–314;
12. Mayer OH, et al. *Pediatr Pulmonol.* 2015;50(5):487–494;
13. McDonald CM, et al. *Neuromuscul Disord.* 2018;28(11):897–909;
14. Batra A, et al. *BMC Cardiovasc Disord.* 2022;22(1):260;
15. Hagenbuch SC, et al. *Am J Cardiol.* 2010;105(10):1451–1455;
16. Powers JM. Approach to the child with anemia. UpToDate, Connor RF (Ed), Wolters Kluwer. Accessed February 18, 2026.

Zeleciment rostudirsen (z-rostudirsen, also known as DYNE-251) is an investigational medicine or otherwise in development and has not been approved as safe or effective by the US FDA, EMA, or any other regulatory authority

Muscular Dystrophy Association Clinical & Scientific Congress (MDA), Orlando, Florida, USA, March 8–11, 2026

Dyne THERAPEUTICS